Revisiting Perampanel: A Literature-Based review on its Role as a first add-on in Focal Seizure Management

Dr Arthik Shetty [1], Dr Prashant Devkare [2], Shruti Dharmadhikari [1], Dr Chintan Khandhedia [1], Dr Amey Mane [1], Dr Suyog Mehta [1]
Affiliation: 1. Sun Pharma Laboratories Ltd, Mumbai; 2. Sun Pharma Industries Ltd, Mumbai

1

Poster presented at 32nd Annual Conference of Indian Academy of Neurology held at Varanasi, India between 29th Oct – 2nd Nov,2025

Introduction

Epilepsy[1]

- India has an estimated epilepsy prevalence ranging from 3.0-11.9 per 1,000 individuals, with approximately 12 million individuals affected nationwide and almost half of them suffering from focal seizures

Challenges faced

- Globally, around 20–30% of persons with epilepsy have drug-resistant epilepsy (DRE)[2]
- An Indian study showed **40-50%** failed to respond to first-line AED monotherapy[3]
- Some monotherapy AEDs cause side effects that lead to treatment withdrawal

Perampanel (AMPA antagonist)

- Perampanel, a highly selective, and non-competitive antagonist of AMPA receptor, approved for adjunctive treatment of partial-onset seizures, with or without secondarily generalized seizures in patients aged ≥12 year
- It's once-daily oral dosing, predictable pharmacology and dose-response (allows gradual up-titration to balance efficacy and neuropsychiatric/coordination AEs), and synergy observed in some combinations make it convenient to introduce as a first add-on[4]

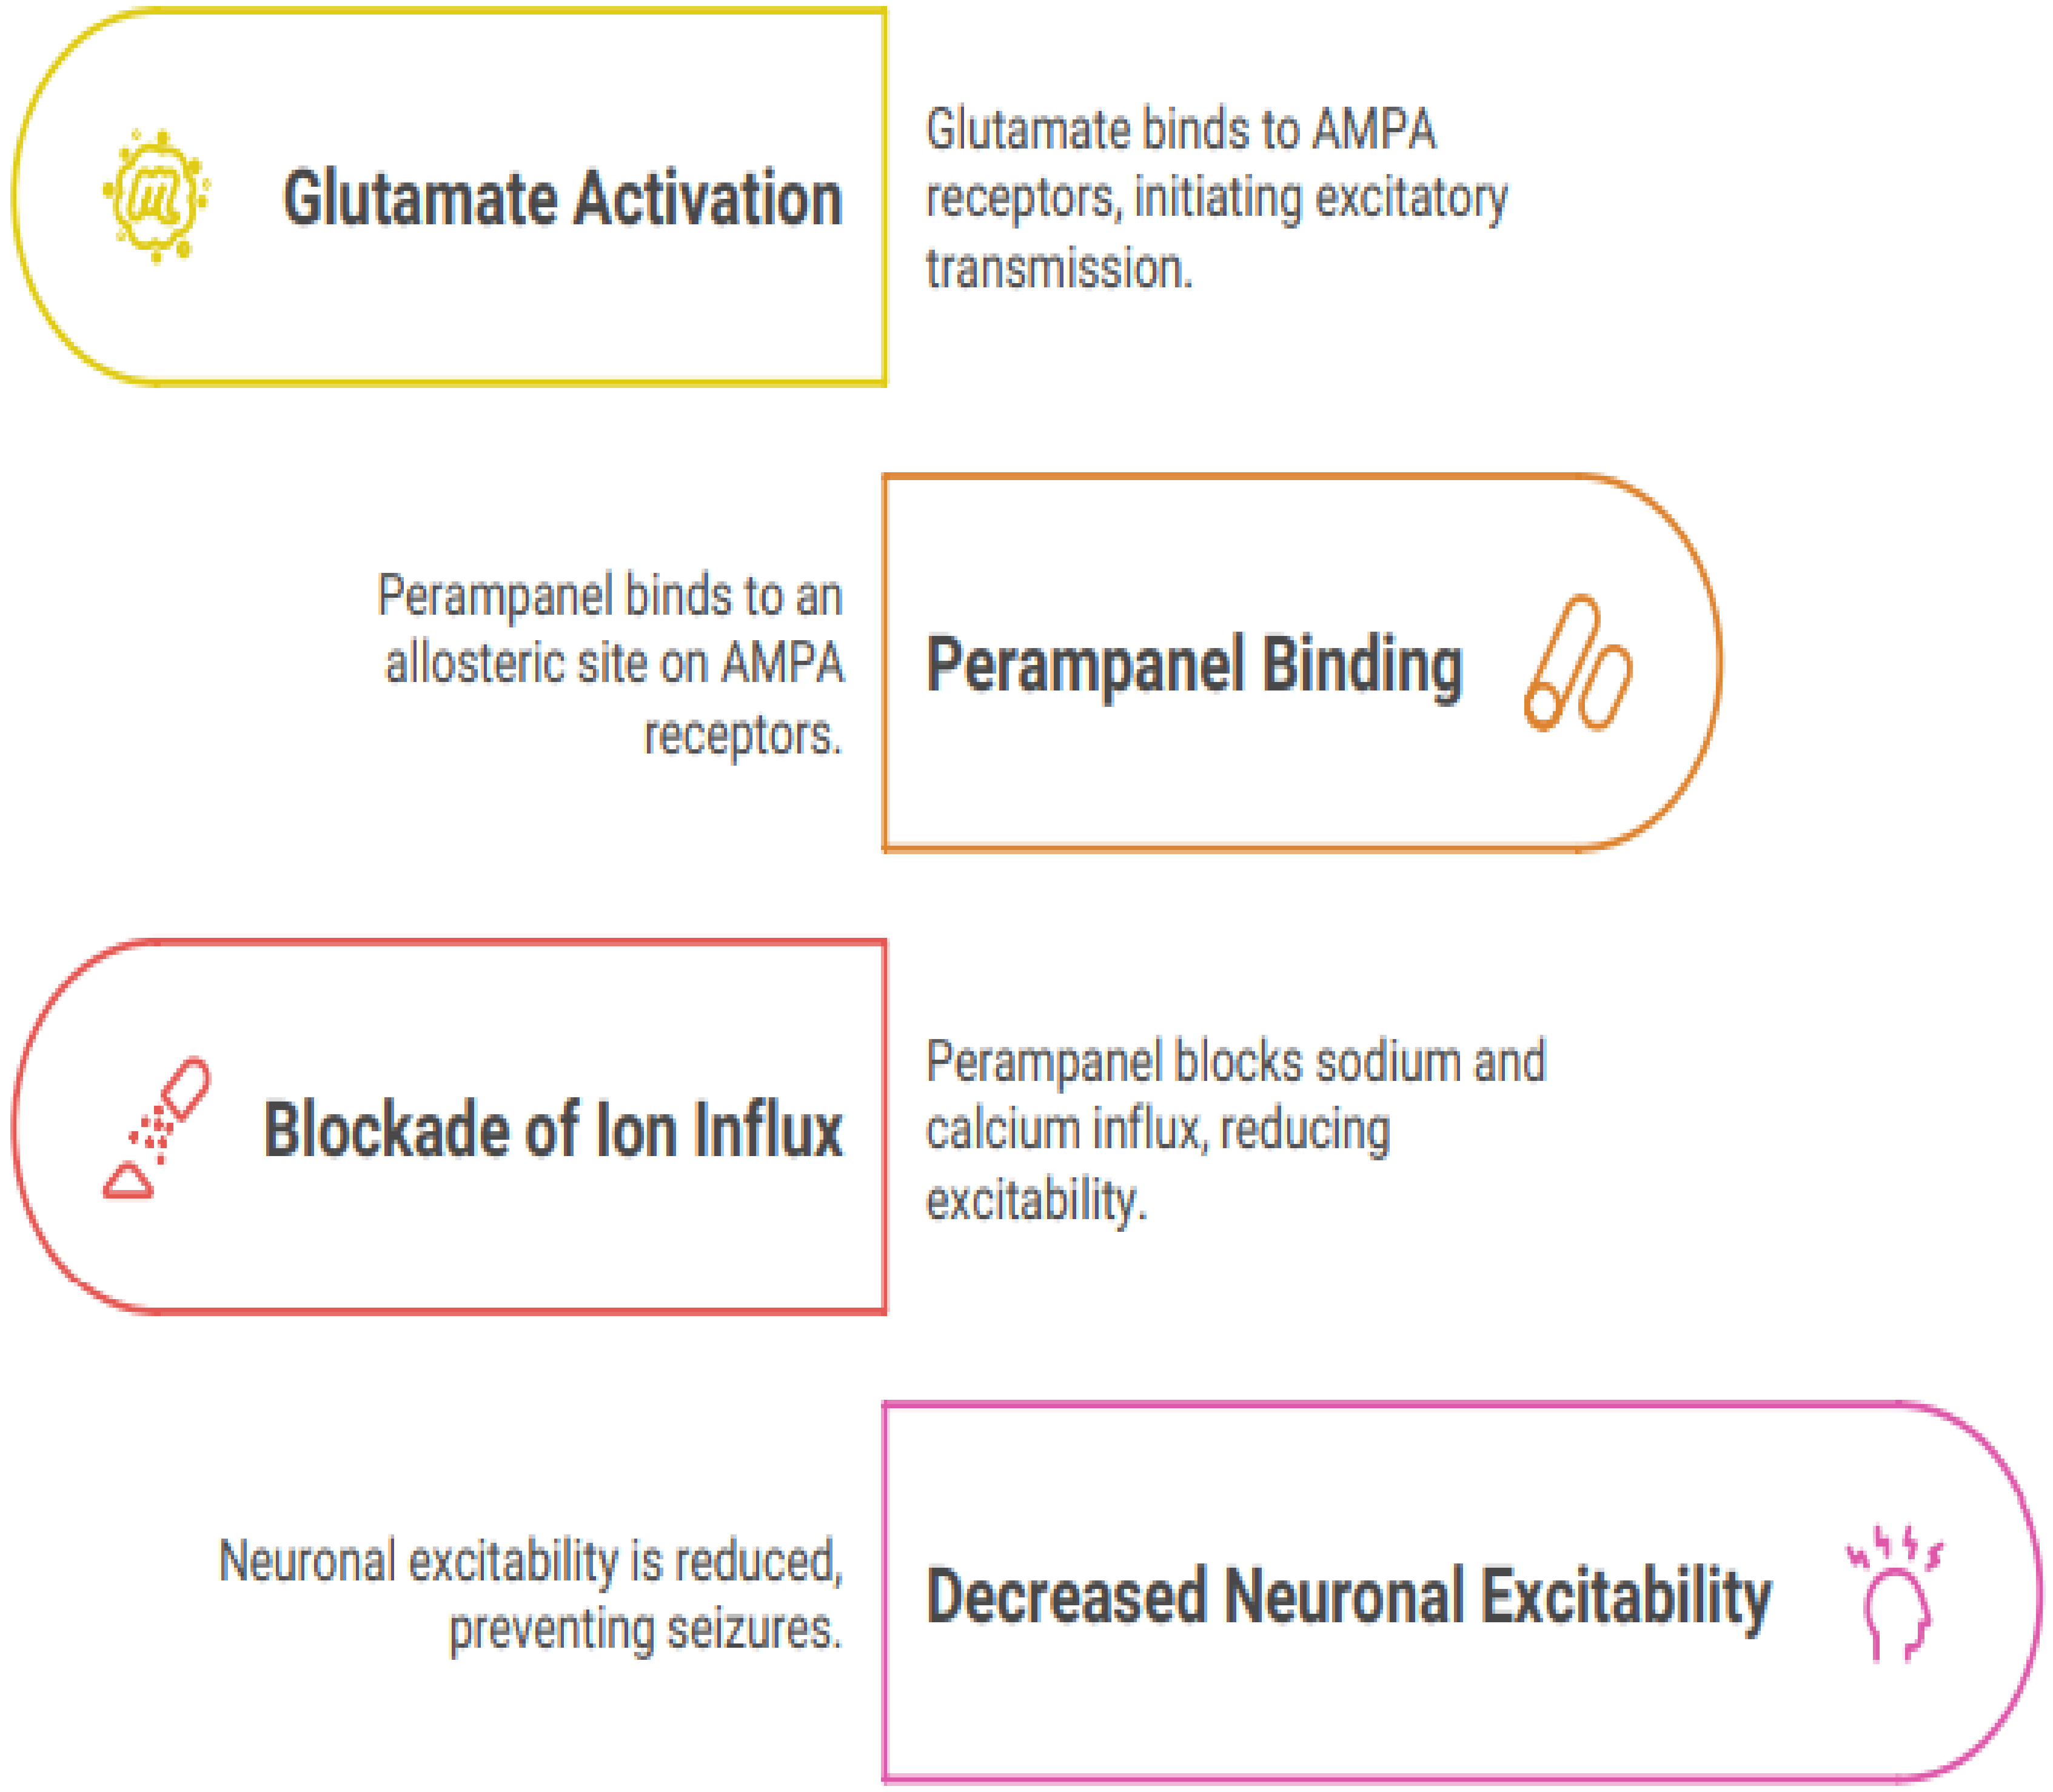
Glutamate Activation
Glutamate binds to AMPA receptors, initiating excitatory transmission.
Perampanel binds to an allosteric site on AMPA receptors.
Perampanel Binding
Blockade of Ion Influx
Perampanel blocks sodium and calcium influx, reducing excitability.
Neuronal excitability is reduced, preventing seizures.
Decreased Neuronal Excitability

1. Ann Indian Acad Neurol. 2015 Jul-Sep;18(3):263-77
2 Clinical Neurology and Neurosurgery,Volume 256,2025,109009,.
3. Rawat, Chitra & Guin, Debleena & Talwar, Puneet & Grover, Sandeep & Baghel, Ruchi & Kushwaha, Suman & Sharma, Sangeeta & Agarwal, Rachna & Bala, Kiran & Srivastava, Achal Kumar & Kukreti, Ritushree. (2018). Clinical predictors of treatment outcome in North Indian patients on antiepileptic drug therapy: A prospective observational study. Neurology India. 66. 1052-1059. 10.4103/0028-3886.237000
4. Hanada, T. (2014). The discovery and development of perampanel for the treatment of epilepsy. Expert Opinion on Drug Discovery, 9(4), 449–458

2

Materials and Methods

Existing literature from 2017-2024 was screened to evaluate the role of perampanel as first add-on in management of partial onset seizures

Primary research articles were obtained from PubMed and Google Scholar.

Keywords included Perampanel, AMPA receptor antagonist, Focal onset seizures, Adjunctive therapy / Add-on therapy , seizure freedom

Aim

To highlight role of perampanel as first add-on in management of partial onset seizures as assessed by seizure freedom rate

3

Results of Perampanel as first add on

	Study name	Baseline	3 months	6 months	1 year	> 1 year
1	Zhang et al (2021) N=56, Observational Prospective	20 (no. of FOS)	Seizure freedom – 8% ≥50% reduction rate – 52%	Seizure freedom – 15.8% ≥50% reduction rate – 55.3%		
2	Seo et al (2019) N=85, Prospective Interventional	85		Seizure freedom – 80% ≥50% reduction rate – 47.1%		
3	Takahashi et al (2019) N=51, Observational Retrospective	13		Seizure freedom – 29% ≥50% reduction rate – 85%	Seizure freedom – 28% ≥50% reduction rate – 80%	
4	Stavropoulos, et al. (2019) N=181, Observational retrospective	134	Seizure freedom – 4.2% ≥50% reduction rate – 16.14%	Seizure freedom – 5.6% ≥50% reduction rate – 19.2%	Seizure freedom – 5.6% ≥50% reduction rate – 20.2%	
5	Santamarina et al. (2020) N=149, Observational Retrospective	113			Seizure freedom – 40.7% ≥50% reduction rate – 82.3%	
6	Gasparini et al (2022) N=503, Observational Restrospective	155	Seizure freedom – 42% ≥50% reduction rate – 66%	Seizure freedom – 50% ≥50% reduction rate – 88%	Seizure freedom – 48% ≥50% reduction rate – 83%	
7	Punia et al (2024) N=54, Phase IV	30			Seizure freedom – 26.7% ≥50% reduction rate – 77.8%	
8	Lee et al (2021) N=106, Extension study	96			Seizure freedom – 43.6% ≥50% reduction rate – 76.4%	Seizure freedom 2 years – 50%, 3 years – 58.7% ≥50% reduction rate 2 years – 79.2%, 3 years – 84.8
9	Kim et al (2022) N=106, Post hoc	79		Seizure freedom – 50.6% ≥50% reduction rate – 83.5%		

Adverse effects

Studies have shown the most common AEs were dizziness (33.8%), somnolence (5.4%), anger (4.1%), and irritability (4.1%). No significant cardiac or metabolic adverse effects were reported.

1. Zhang R, Qiao S, Fang X, Wang K, Shi Y, Du Q, Yang T and Liu X (2021) Efficacy and Tolerability of Perampanel as Adjunctive Therapy in Chinese Patients With Focal-Onset Seizures: An Observational, Prospective Study. Front. Neurol. 12:731566. .
2. Takahashi S, Shimizu K, Inaji M, Hashimoto S, Yamamoto S, Maehara T. Effectiveness of perampanel as a first add-on antiepileptic drug for the treatment of partial epilepsy. Epilepsy Behav. 2019 Nov;100(Pt A):106492
3. Stavropoulos I, Louden W, Queally C, Adcock J, Tristram M, Neale M, Moran N, Flores L L, Nashef L, Richardson MP, Bell C, Slaght S, Aram J, Rayner N, Powell R, Mead A, Sen A, Elwes R. Perampanel for the treatment of epilepsy; Longitudinal actuarial analysis and dose responses based on monthly outcomes. Seizure. 2019 Jul;69:125-132.
4. Punia V, Klein P, Mihaylova T, Biton V, Samad O, Ngo LY, Kumar D, Malhotra M. Perampanel as monotherapy or first adjunctive therapy in pediatric and adult patients with epilepsy: the first United States-based phase IV open-label ELEVATE study. J Neurol. 2024 Jul;271(7):4587-4598.
5. Im K, Lee SA, Kim JH, Kim DW, Lee SK, Seo DW, Lee JW. Long-term efficacy and safety of perampanel as a first add-on therapy in patients with focal epilepsy: Three-year extension study. Epilepsy Behav. 2021 Dec;125:108407
6. . Kim JH, Kim DW, Lee SK, Seo DW, Lee JW, Park HJ, Lee SA. First add-on perampanel for focal-onset seizures: An open-label, prospective study. Acta Neurol Scand. 2020 Feb;141(2):132-140

4

Conclusion

- Evidence has shown perampanel being reliable option in focal seizure management with seizure freedom ranging from 8%- 58% and ≥50% reduction rate from 16%-84% with duration between 3 months- 3 years .
- A desirable feature of perampanel which is relevant to add-on use is its unique mechanism, which facilitates use in combination with any antiseizure medication.
- No significant cardiac or metabolic adverse effects were reported, supporting long-term safety.
- It’s role as add-on is useful in polytherapy settings due to low-interaction potential.
- Overall, perampanel showed significant efficacy and safety as first add-on in patients with focal seizures

5

Poster presented at 32nd Annual Conference of Indian Academy of Neurology held at Varanasi, India between 29th Oct – 2nd Nov,2025